The Italian Multiple Sclerosis And Related Disorders Register
Registro Italiano Sclerosi Multipla E Patologie Correlate (RISM)

Distribution of MS Clinical Centers, the Network of Research Assistants and MS cases collected in RISM by Italian Regions

RISM
The Italian Multiple Sclerosis and Related Disorders Register

NETWORK OF RESEARCH ASSISTANTS

26 Research Assistants (RAs) are working in 15 Regions to support more than 130 MS Clinical Centers.

The activities of the RAs include:
- supporting the start-up phase of the project at the MS Centers
- ensuring a standardized collection of high-quality data
- playing a key role in communication between the RISM infrastructures and the MS Centers.

89,746 MS cases

26 Research Assistants

189 MS Clinical Centers have joined RISM

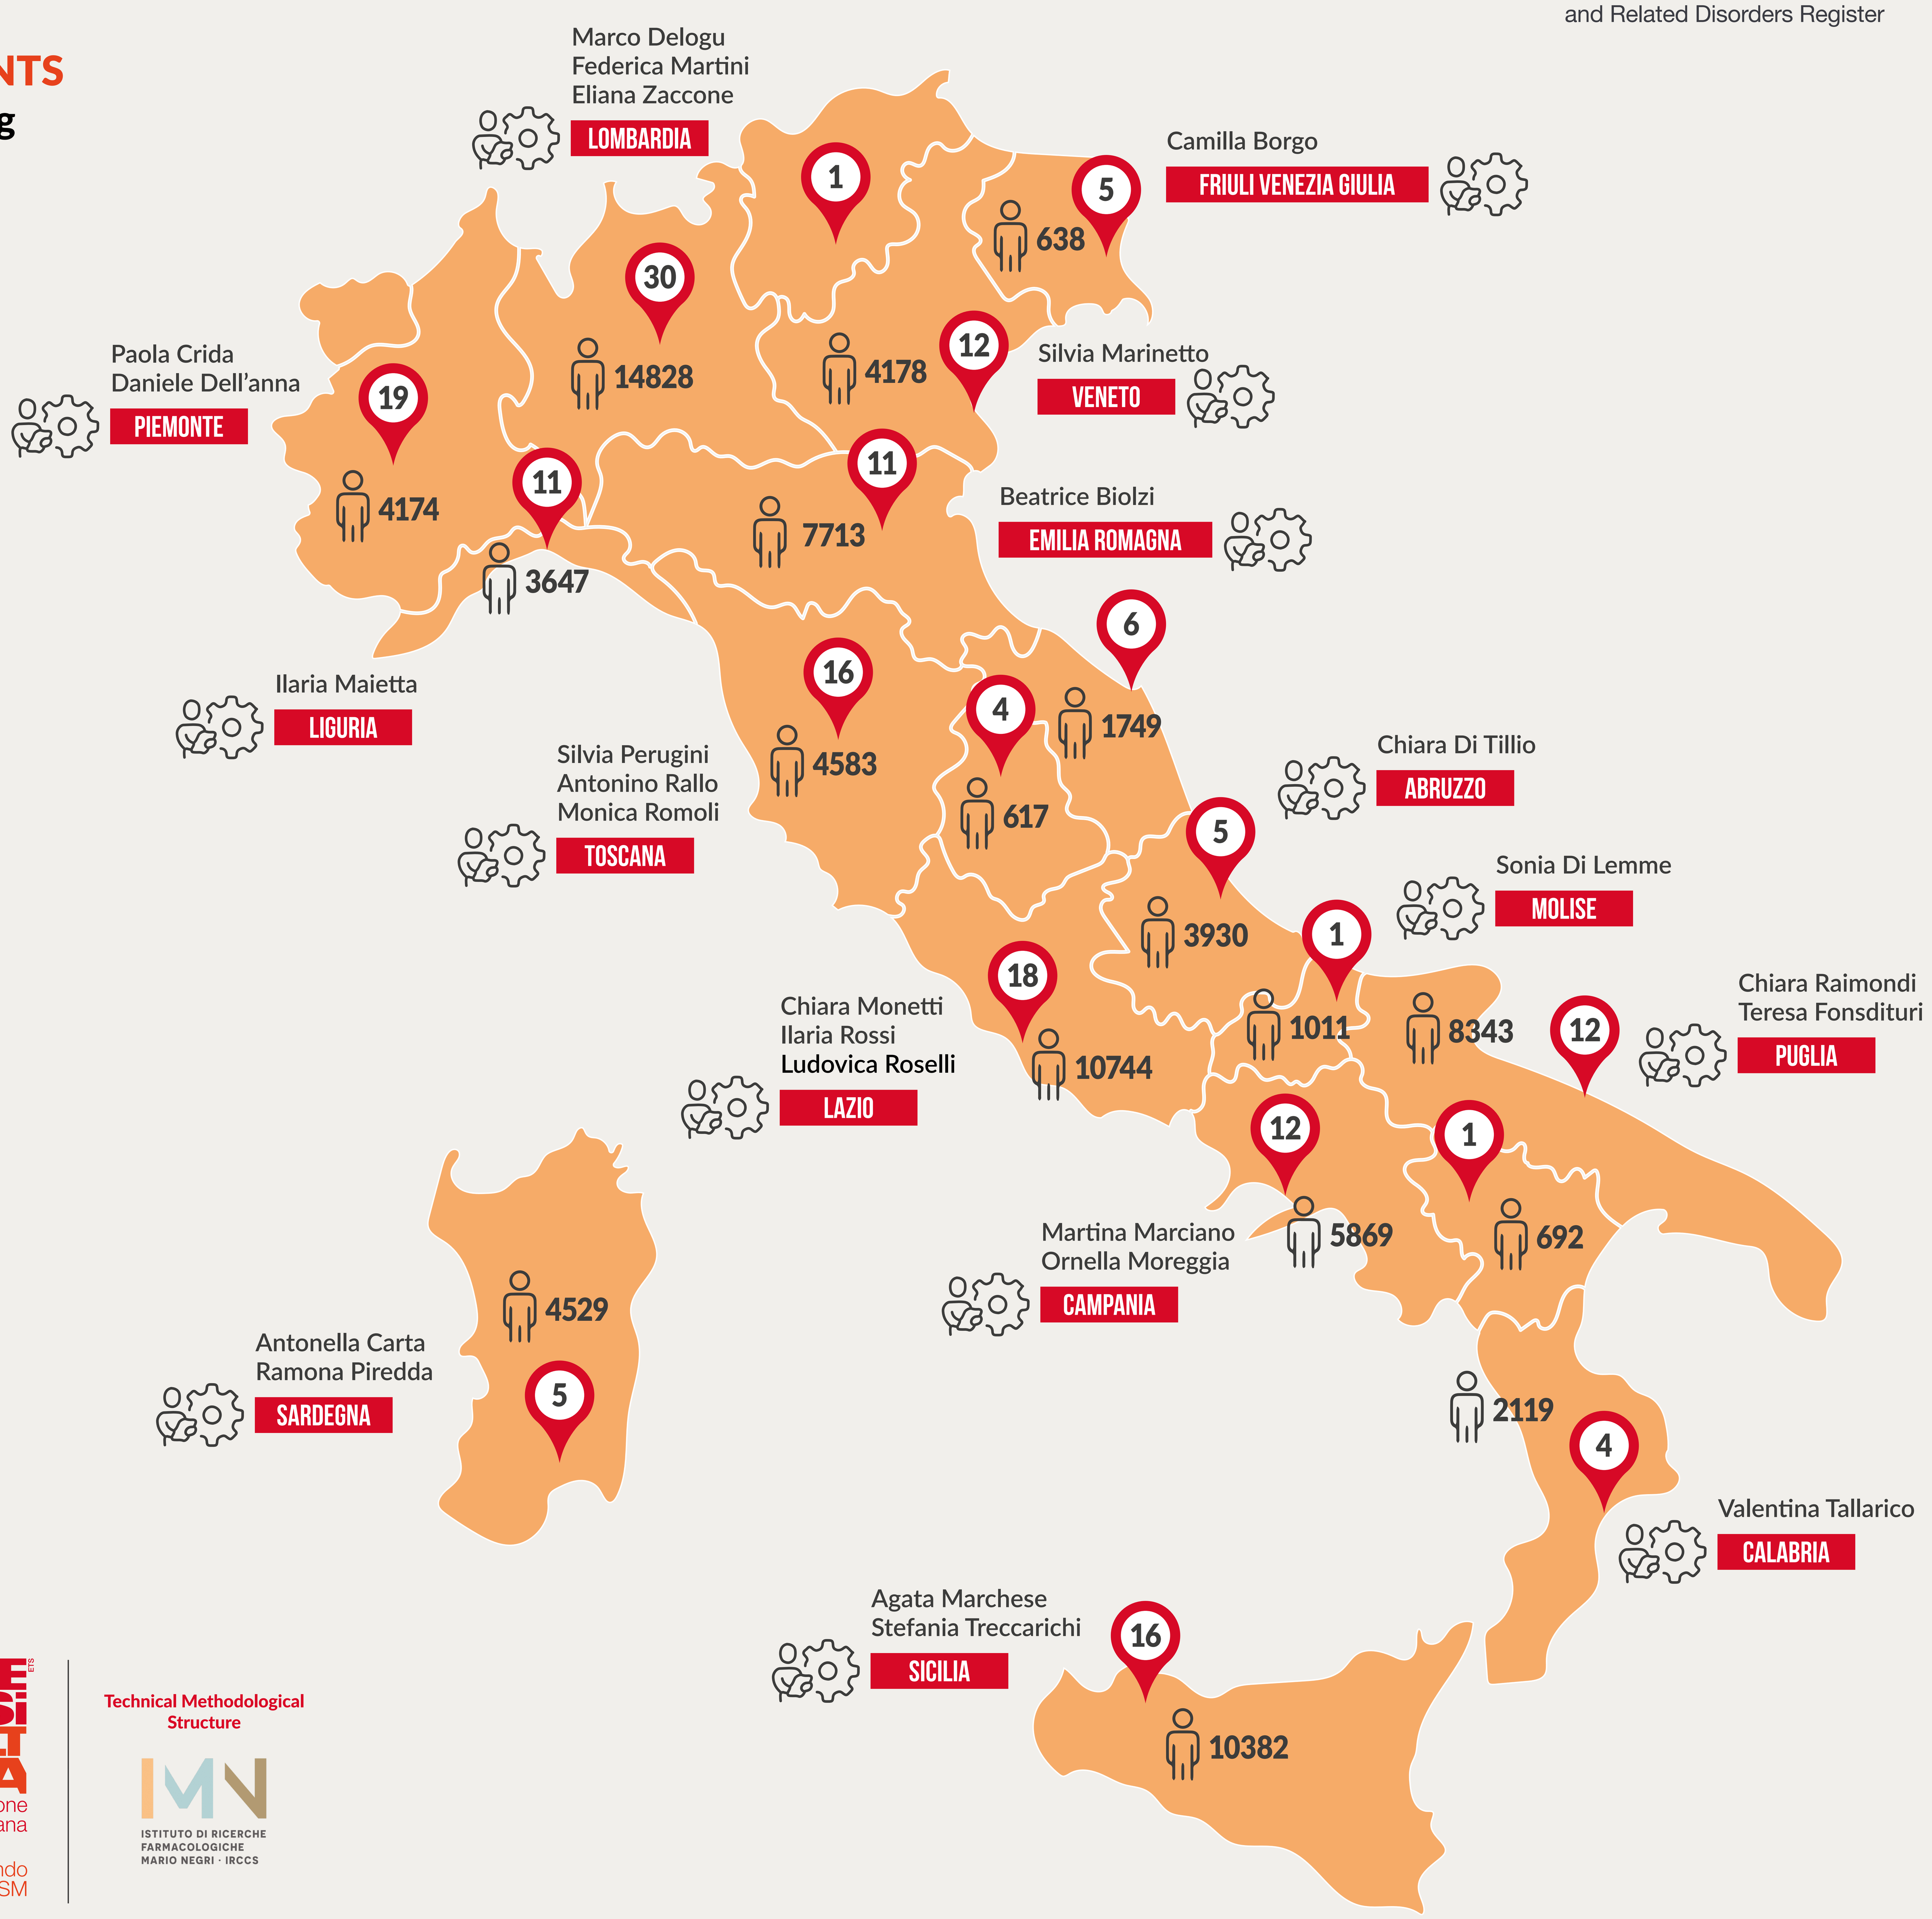

Marco Delogu
Federica Martini
Eliana Zaccone
LOMBARDIA
30
14828
Camilla Borgo
FRIULI VENEZIA GIULIA
5
638
1
Silvia Marinetto
VENETO
12
4178
Paola Crida
Daniele Dell'anna
PIEMONTE
19
4174
11
3647
Beatrice Biolzi
EMILIA ROMAGNA
11
7713
Ilaria Maietta
LIGURIA
Silvia Perugini
Antonino Rallo
Monica Romoli
TOSCANA
16
4583
6
1749
4
617
Chiara Di Tillio
ABRUZZO
5
3930
Sonia Di Lemme
MOLISE
1
1011
8343
Chiara Raimondi
Teresa Fonsdituri
PUGLIA
12
Chiara Monetti
Ilaria Rossi
Ludovica Roselli
LAZIO
18
10744
12
5869
1
692
Martina Marciano
Ornella Moreggia
CAMPANIA
2119
4
Valentina Tallarico
CALABRIA
Antonella Carta
Ramona Piredda
SARDEGNA
4529
5
Agata Marchese
Stefania Treccarichi
SICILIA
16
10382

www.registroitalianosm.it

UNIVERSITÀ DEGLI STUDI DI BARI ALDO MORO

NETWORK OF THE ITALIAN MS CLINICAL CENTERS

SCLEROSI MULTIPLA fondazione italiana ETS
un mondo libero dalla SM

Technical Methodological Structure
ISTITUTO DI RICERCHE FARMACOLOGICHE MARIO NEGRI · IRCCS

Unconditionally supported by:

ALEXION AstraZeneca Rare Disease · Bristol Myers Squibb · MERCK · NOVARTIS Reimagining Medicine · Roche · SANDOZ · VIATRIS

List of participating MS Clinical Centers divided by Italian Regions
Clinical Center, Principal Investigator, Starting Year of Participation

ABRUZZO
UOC Neurologia & Stroke Unit - Ospedale Civile G. Mazzini di Teramo - Villa Mosca, Samanta Sciamanna, 2024
Divisione di Neurologia - Ospedale Civile San Filippo e Nicola, Federica De Santis, 2024
Centro SM - UO Neurologia - Casa di Cura Villa Serena, Francesco D'Andrea, 2015
Centro Sclerosi Multipla - Clinica Neurologica Policlinico SS. Annunziata, Stefano Sensi, 2016
Centro Malattie Demielinizzanti - Clinica Neurologica Ospedale San Salvatore, Rocco Totaro, 2016

BASILICATA
Centro Sclerosi Multipla - PO Madonna delle Grazie, Maria Gabriella Coniglio, 2015

CALABRIA
Ambulatorio SM - Grande Ospedale Metropolitano Bianchi Melacrino Morelli, Umberto Aguglia, 2015
Centro SM - Divisione di Neurologia - Ospedale dell'Annunziata, Maria Trotta, 2021
Centro Sclerosi Multipla - Policlinico Universitario - Campus Germaneto, Paola Valentino, 2015
UO di Neurologia - Centro SM - ASP di Cosenza, Roberto Bruno Bossio, 2016

CAMPANIA
IRCCS SYNLAB SDN SpA, Carlo Cavaliere, 2022
Centro Regionale di Diagnosi e Terapia della Sclerosi Multipla, Giorgia Teresa Maniscalco, 2016
Centro Regionale Sclerosi Multipla - Unità Operativa Semplice - AOU Policlinico Federico II, Vincenzo Brescia Morra, 2016
Centro Clinico per la Sclerosi Multipla - II Clinica Neurologica - II Università di Napoli, Giacomo Lus, 2015
Centro Sclerosi Multipla - UOC Neurologia - AORN San Giuseppe Moscati, Daniele Spitaleri, 2015
SC di Neurologia - Dipartimento di Neuroscienze e Riabilitazione - AORN Santobono Pausilipon, Marida Della Corte, 2020
Dipartimento di Scienze Mediche e Chirurgiche Avanzate - Università della Luigi Vanvitelli, Antonio Gallo, 2015
Divisione Neurologia - Azienda Ospedaliera San Giovanni di Dio e Ruggi, Paolo Barone, 2018
UOC Neurologia - UOS Neurofisiologia - Dipartimento di Neuroscienze, Scienze Riproduttive ed Odontostomatologiche, Università di Napoli Federico II, Rosa Iodice, 2015
Centro SM - UOC di Neurologia - Ospedale San Paolo - ASL Napoli 1 Centro, Leonardo Sinisi, 2015
UO di Neurologia - Ospedale Maria SS. Addolorata - Centro Diagnosi e Terapia Sclerosi Multipla, Vincenzo Busillo, 2015
Centro Sclerosi Multipla - PO San Giuseppe Moscati, Nicola Capasso, 2019

EMILIA-ROMAGNA
Ambulatorio Sclerosi Multipla della UO di Neurologia - Ospedale Morgagni Pierantoni - AUSL della Romagna, Silvia Strumia, 2015
Centro SM UOC Neurologia - Ospedale di Vaio - AUSL PR, Ilaria Pesci, 2015
UOC di Neurologia - Ospedale Ramazzini, Mario Santangelo, 2016
Dipartimento di Neuroscienze - Ospedale Civile di Baggiovara - Azienda Ospedaliero-Universitaria, Diana Ferraro, 2015
Azienda Unità Sanitaria Locale di Piacenza - Ospedale Guglielmo da Saliceto - UOC Neurologia, Paolo Immovilli, 2019
Dipartimento di Neuroscienze - Centro Sclerosi Multipla - UO Neurologia - Ospedale S. Maria delle Croci - AUSL Romagna, Matteo Foschi, 2021
Centro Sclerosi Multipla - UOC di Neurologia - Dipartimento di Medicina Generale e Specialistica - AOU di Parma, Franco Granella, 2015
IRCCS Istituto delle Scienze Neurologiche di Bologna - UOSI Riabilitazione Sclerosi Multipla, Alessandra Lugaresi, 2015
Centro SM - UOC Neurologia - Arcispedale Santa Maria Nuova - AUSL Reggio Emilia, Sara Montepietra, 2015
Centro di Servizio e Ricerca sulla Sclerosi Multipla AOU di Ferrara, Maura Pugliatti, 2015
Centro Sclerosi Multipla - UO di Neurologia - Ospedale Bufalini, Luca Mancinelli, 2016

FRIULI-VENEZIA GIULIA
SOC Neurologia - Day Hospital - ASUIUD PO S. Maria Della Misericordia, Daniela Cargnelutti, 2015
Divisione Neurologica - Ospedale Civile Santa Maria degli Angeli, Laura Locatelli, 2022
Centro di Studio e cura Sclerosi Multipla ASUGI - Ospedale di Cattinara, Antonio Bosco, 2022
Struttura Complessa di Neuropsichiatria Infantile - IRCCS Materno Infantile Burlo Garofolo, Marco Carrozzi, 2020
Centro SM Clinica Neurologica Udine, Mariarosaria Valente, 2023

LAZIO
Centro Clinico Sclerosi Multipla - Ospedale Fatebenefratelli San Pietro, Marco Peresson, 2015
Centro Sclerosi Multipla - AO S. Camillo Forlanini, Carla Tortorella, 2015
Centro Sclerosi Multipla - AO San Giovanni Addolorata, Carlo Piantadosi, 2015
CENTERS Centro Neurologico Terapie Sperimentali - Sapienza Università di Roma - AO S. Andrea, Marco Salvetti, 2015
Centro SM - Policlinico S. Andrea - Università Sapienza, Carlo Pozzilli, 2015
Centro SM - Fondazione Santa Lucia IRCCS, Maria Grazia Grasso, 2015
Centro SM - Ospedale Spaziani, Fabiana Marinelli, 2021
Centro per la diagnosi e la cura delle malattie infiammatorie demielinizzanti in eta' pediatrica - Ospedale Bambino Gesù, Massimiliano Valeriani, 2017
UOSD Sclerosi Multipla - Dipartimento Medicina dei Sistemi - Università Tor Vergata, Girolama Alessandra Marfia, 2015
UO Sclerosi Multipla - Fondazione Policlinico Universitario A. Gemelli IRCCS - Università Cattolica del Sacro Cuore, Massimiliano Mirabella, 2015
Dipartimento di Neuroscienze Umane - Unità di Neuropsichiatria Infantile - Università La Sapienza, Vincenzo Leuzzi, 2020
Centro Sclerosi Multipla - Ospedale Belcolle, Nicola Falcone, 2019
Centro Regionale Diagnosi e Cura Sclerosi Multipla e Malattie Demielinizzanti - Ospedale Sant'Eugenio, Daniela De Pascalis, 2015
Servizio di Neurologia - Ospedale Santa Maria Goretti, Francesco Sica, 2021
Centro Sclerosi Multipla - UO di Neurologia - Ospedale San Camillo De Lellis, Steno Rinalduzzi, 2015
Centro Sclerosi Multipla - AOU del Policlinico Umberto I - Università Sapienza, Antonella Conte, 2017
Centro SM - UOC di Neurologia - Policlinico Universitario Campus Bio-Medico, Vincenzo Di Lazzaro, 2015
Centro SM - PO San Filippo Neri - ASL Roma 1, Elisabetta Ferraro, 2015

LIGURIA
Centro Sclerosi Multipla - Divisione di Neurologia - Ospedale San Paolo, Matteo Pizzorno, 2015
Centro SM Pietra Ligure SC Neurologia - Ospedale Santa Corona, Tiziana Tassinari, 2015
Ambulatorio Sclerosi Multipla - Neurologia ASL 4 Chiavarese, Giuseppe Trivelli, 2017
Ambulatorio Sclerosi Multipla - EO Ospedali Galliera, Simonetta Venturi, 2015
Centro Sclerosi Multipla e Malattie Demielinizzanti - Ospedale Civile S. Andrea - USL 5, Loredana Petrucci, 2015
UO Neuropsichiatria Infantile - Dip. Neuroscienze Mediche e Chirurgiche e Riabilitazione - Continuità delle Cure - IRCCS Gaslini, Maria Margherita Mancardi, 2020
SC Neurologia - Centro Sclerosi Multipla - Ospedale Padre Antero Micone - ASL3 Genovese, Paola Gazzola, 2015
Centro per lo Studio e la Cura della Sclerosi Multipla e Malattie Demielinizzanti - Dipartimento di Neuroscienze, Riabilitazione, Oftalmologia, Genetica e Scienze Materno-Infantili - Clinica Neurologica - Ospedale Policlinico San Martino (DiNOGMI), Matilde Inglese, 2015
UO Neurologia - Dipartimento di Neuroscienze e Organi di Senso - Ospedale Policlinico San Martino, Giuseppe Ribizzi, 2015
Ambulatorio Malattie Demielinizzanti - OC Imperia, Maria Teresa Rilla, 2022
Servizio di Riabilitazione AISM Liguria, Giampaolo Brichetto, 2017

LOMBARDIA
SC Neurologia Pediatrica - Centro Sclerosi Multipla Pediatrica - Ospedale dei Bambini Buzzi, Stefania Maria Bova, 2020
Neurologia ad Indirizzo Neuroimmunologico - Centro Sclerosi Multipla - ASST della Valle Olona - Ospedale di Gallarate, Pietro Annovazzi, 2015
ASST della Valle Olona - PO di Saronno - Ambulatorio Sclerosi Multipla, Davide Nasuelli, 2016
Centro di Riferimento Regionale per la Sclerosi Multipla - Spedali Civili Brescia - PO Montichiari, Nicola De Rossi, 2023
Centro di Neuroimmunologia - Fondazione IRCCS San Gerardo dei Tintori, Maura Frigo, 2015
Fondazione IRCCS Istituto Neurologico Carlo Besta, Valentina Torri Clerici, 2015
Centro SM - Fondazione Don Carlo Gnocchi IRCCS, Marco Rovaris, 2015
ASST Grande Ospedale Metropolitano Niguarda - Centro SM, Alessandra Protti, 2015
Azienda Socio Sanitaria Territoriale (ASST) della Franciacorta, Vincenzo Sidoti, 2017
Centro SM - Dipartimento di Neuroscienze - ASST Ovest Milanese, Serena Leva, 2015
Centro Sclerosi Multipla - Ospedale di Desio, Ignazio Santilli, 2023
Centro Sclerosi Multipla - Fondazione IRCCS Ca' Granda Ospedale Maggiore Policlinico, Elio Scarpini, 2016
Centro SM - Ambulatorio Malattie Demielinizzanti - Ospedale di Circolo e Fondazione Macchi, Paola Banfi, 2015
Centro Sclerosi Multipla - IRCCS Fondazione Mondino, Roberto Bergamaschi, 2015
Centro ad Alta Specializzazione per la diagnosi e la cura della Sclerosi Multipla - Ospedale Valduce, Raffaella Clerici, 2015
Centro Provinciale Sclerosi Multipla - ASST Papa Giovanni XXIII, Marta Zaffira Conti, 2015
Neuroimmunologia - Centro Provinciale per la diagnosi e terapia della Sclerosi Multipla - ASST Crema, Maria Teresa Ferrò, 2015
SC Neurologia - ASST Valcamonica - Ospedale di Esine, Marinella Turla, 2017
Centro SM - ASST Rhodense, Marco Ronzoni, 2016
Centro Sclerosi Multipla - Divisione di Neurologia - Azienda Ospedaliera A. Manzoni, Roberto Balgera, 2015
Dipartimento di Neuroscienze Pediatriche - Fondazione IRCCS Istituto Neurologico C. Besta, Alessandra Tozzo, 2020
Centro Sclerosi Multipla - UO Neurologia - ASST Fatebenefratelli Sacco - PO Luigi Sacco, Pierluigi Bertora, 2015
UO Neurologia - Ospedale Maggiore, Silvia Fermi, 2018
Centro Sclerosi Multipla - Ospedale San Carlo - ASST Santi Paolo e Carlo, Giuseppe Santuccio, 2015
IRCCS Fondazione Mondino - UO Neuropsichiatria infantile, Angela Berardinelli, 2020
Centro SM UO Neurologia ASST Lariana Ospedale S. Anna, Nerina Mascoli, 2015
Centro Sclerosi Multipla - Unità di Neurologia - IRCCS Ospedale San Raffaele, Massimo Filippi, 2016
Azienda Socio Sanitaria Territoriale (ASST) della Valtellina e Alto Lario - Sedi di Sondrio e Sondalo - Reparto di Neurologia, Alessandro Leone, 2015
Centro Sclerosi Multipla Casa di Cura Igea, Giancarlo Comi, 2022
Istituto Clinico Humanitas - HUNIMED, Giuseppe Liberatore, 2022

MARCHE
Clinica Neurologica delle Marche - AOU delle Raffaella Cerqua, 2015
Ambulatorio Sclerosi Multipla - UO di Neurologia AV5 - Ospedale Civile Madonna del Soccorso, Cristina Paci, 2015
Ambulatorio Sclerosi Multipla - UO Neurologia AV5 - Ospedale C. e G. Mazzoni, Gabriella Cacchiò, 2015
Centro Sclerosi Multipla - c/o UOC Neurologia - Ospedale di Macerata, Elisabetta Cartechini, 2015
UOC Neurologia - Ospedale Civile di Senigallia, Giorgia Jorio, 2024
UO Neurologia, Azienda Ospedaliera Marche Nord, Centro SM, Cristiana Taus, 2022

MOLISE
Centro Sclerosi Multipla - IRCCS Neuromed, Paolo Bellantonio, 2015

PIEMONTE
Divisione di Neurologia - Ospedale Civile, Carlotta Chiavazza, 2022
Centro Sclerosi Multipla - SC Neurologia - AO S. Croce e Carle, Marco Capobianco, 2015
Centro SM - Ospedale Regina Montis Regalis, Gabriella Turano, 2017
Centro Sclerosi Multipla - Clinica Neurologica - Dipartimento di Medicina Traslazionale - Università del Piemonte Orientale, Roberto Cantello, 2017
Dipartimento di Riabilitazione - CRRF Mons. Luigi Novarese, Claudio Solaro, 2018
Centro SM ASL CN2 Alba-Bra, Sara Giacone, 2023
Centro Sclerosi Multipla - Divisione di Neurologia - Ospedale Maria Vittoria, Daniele Imperiale, 2016
Centro di riferimento Regionale SM (CRESM) - SCDO Neurologia - AOU San Luigi Gonzaga, Alessia Di Sapio, 2015
Divisione di Neurologia - Ospedale Martini, Valeria Studer, 2023
Divisione di Neurologia - Ospedale SS. Annunziata, Maria Roberta Bongioanni, 2023
Istituto Auxologico Italiano IRCCS - Istituto Scientifico Ospedale S. Giuseppe e Ambulatorio, Andrea Mauro Brioschi, 2017
Servizio Universitario Sclerosi Multipla - Dip. di Neuroscienze Rita Levi Montalcini dell'Università di Torino - Unità Operativa Complessa Neurologia 2 dell'AOU Città della Salute e della Scienza di Torino, Marco Bozzali, 2020
Centro Sclerosi Multipla - Divisione Di Neurologia - Ospedale Civile - ASL 4, Giulia De Rosa, 2015
AOU SS Antonio e Biagio e Cesare Arrigo, Francesco Passantino, 2018
ASLTO4 - Neurologia - Ospedale di Chivasso, Claudio Geda, 2015
Centro Clinico delle Malattie Demielinizzanti dell'ASL di Biella - Ospedale degli Infermi, Lorenzo Capone, 2015
Struttura Complessa a Direzione Ospedaliera (SCDO) Neurologia 1 - AOU San Luigi Gonzaga, Marinella Clerico, 2015
Dipartimento di Scienza della Sanità Pubblica e Pediatriche SCDU Neuropsichiatria Infantile AOCittà della Salute e della Scienza della Città di Torino Presidio Ospedale Infantile Regina Margherita, Carlotta Canavese, 2022
Centro SM - Neurologia 1U - AOU Città della Salute e della Scienza di Torino, Paola Cavalla, 2017

PUGLIA
Centro SM - DiBraiN - Dipartimento di Biomedicina Traslazionale e Neuroscienze - Università di Bari, Pietro Iaffaldano, 2017
Divisione di Neurologia - Ospedale Vito Fazzi, Francesca De Robertis, 2017
Centro Sclerosi Multipla - UOC Neurologia - Ospedale A. Perrino, Augusto Rini, 2015
Centro SM c/o UO di Neurologia - PO Dimiccoli, Imma Plasmati, 2015
Centro Malattie Demielinizzanti - Ospedale Generale Regionale F. Miulli, Maurizia Gatto, 2015
Ospedale Civile Di Venere, Mariangela D'Onghia, 2021
Centro Interdipartimentale per le Malattie Demielinizzanti - SC Neurologia Universitaria - AOU Policlinico Foggia, Carlo Avolio, 2015
Centro Sclerosi Multipla - UOC di Neurologia Giulio Coppola - Ospedale Civile F. Ferrari, Roberto De Masi, 2023
Divisione di Neurologia - Ospedale SS Annunziata, Rossana Sgobio, 2022
Centro Sclerosi Multipla UOC di Neurologia - Ospedale Della Murgia Fabio Perinei, Ardito Bonaventura, 2015
UOC Neuropsichiatria Infantile Universitaria - Policlinico di Bari, Marta Simone, 2020
Centro SM UO Neurologia - Fondazione IRCCS Casa Sollievo della Sofferenza, Maurizio A. Leone, 2015

SARDEGNA
Centro Prescrittore Sclerosi Multipla - UO di Neurologia - Presidio Ospedaliero di Ozieri - ASL N. 1 Sassari, Antonello Pala, 2015
UOC Neuropsichiatria Infantile - AOU di Sassari, Stefano Sotgiu, 2020
Centro diagnosi, cura e ricerca per Sclerosi Multipla - Ospedale S. Francesco - USL 3, Maria Luisa Piras, 2016
Centro Regionale per la diagnosi e la cura della Sclerosi Multipla ASL8 - PO Binaghi, Eleonora Cocco, 2016
SS Sclerosi Multipla - SC Clinica Neurologica AOU Sassari, Roberto Zarbo, 2015

SICILIA
Centro Sclerosi Multipla - c/o UOC di Neurologia - AO Papardo, Sonia Milone, 2015
Centro Sclerosi Multipla - UOC Neurologia - Ospedale Garibaldi Centro, Francesca Matta, 2015
Centro Sclerosi Multipla - Ospedale ARNAS Civico Di Cristina Benfratelli, Salvatore Cottone, 2020
Divisione di Neurologia - Azienda Ospedaliera S. Elia, Michele Maria Vecchio, 2019
UOC Neurologia - Azienda Ospedaliera Cannizzaro, Davide Maimone, 2023
Centro Neurolesi Bonino Pulejo IRCCS, Edoardo Sessa, 2017
Dipartimento di Radiologia, Diagnostica, Interventistica e Stroke - Azienda Ospedaliera Universitaria Policlinico Paolo Giaccone, Giuseppe Salemi, 2015
Centro Sclerosi Multipla - AOU Policlinico Martino, Maria Buccafusca, 2015
UOC Neurologia con Stroke Unit - Presidio Ospedaliero S. Antonio Abate, Simona Alessi, 2024
Fondazione Istituto G. Giglio - Centro SM, Luigi M. E. Grimaldi, 2015
Centro Sclerosi Multipla - UOC di Neurologia con Stroke Unit - AOOR Villa Sofia-Cervello, Sabrina Realmuto, 2015
Ospedale E. Muscatello, Sebastiano Bucello, 2019
UOC di Neuropsichiatria Infantile Centro Regionale Sclerosi Multipla e Malattie Demielinizzanti Ospedale Pediatrico Giovanni Di Cristina dell'Azienda ARNAS Civico - Di Cristina Benfratelli, Giuseppe Santangelo, 2017
SC Provinciale di Neurologia - ASP Ragusa - PO R. Guzzardi, Antonello Giordano, 2015
Centro Sclerosi Multipla - AOL Policlinico Vittorio Emanuele - Università di Catania, Francesco Patti, 2015
UOC di Neuropsichiatria Infantile - Policlinico Universitario G. Martino, Gabriella Di Rosa, 2020

TOSCANA
Centro SM PO di Grosseto - Azienda USL Toscana Sud Est - Ospedale Misericordia, Katrin Plewnia, 2015
Ambulatorio SM - UOC Clinica Neurologica e Malattie Neurometaboliche - AOU Senese, Nicola De Stefano, 2019
Ambulatorio SM - UOSD Neurologia - Ospedale Valdarno S. Maria alla Gruccia, Francesca Rosini, 2015
Centro Malattie Demielinizzanti - UO Neurologia - Nuovo Ospedale delle Apuane (NOA), Isabella Righini, 2015
UOC Neurologia e Neurofisiologia Clinica - Università degli Studi di Siena, Monica Ulivelli, 2015
Dipartimento NEUROFARBA - Sezione Neuroscienze - Università degli Studi di Firenze - Centro SM SODc Riabilitazione Neurologica - AOU Careggi, Maria Pia Amato, 2015
Centro Aziendale SM - UO Neurologia - Ospedale S. Donato, Benedetta Calchetti, 2020
Ospedale di Prato - Centro per la Sclerosi Multipla - Unità Operativa di Neurologia, Mario Falcini, 2015
Ambulatorio Sclerosi Multipla - Unità Operativa di Neurologia e Neurofisiopatologia - Spedali Riuniti, Cristina Fioretti, 2015
Centro Malattie Disimmuni del SNC e SNP Ospedale San Luca, Patrizia Alessandra Maritato, 2024
Ambulatorio Malattie Demielinizzanti - UOC Neurologia - Ospedale Lotti, Chiara Pecori, 2015
UOC Neurologia Pediatrica AOU Meyer, Federico Melani, 2020
Centro di Riferimento Regionale per il Trattamento della Sclerosi Multipla - SOD Neurologia II - AOU Careggi, Luca Massacesi, 2020
Ospedale San Jacopo - Ambulatorio Malattie Demielinizzanti - Divisione Neurologia, Anna Luisa Ancona, 2020
UO Neurologia - Ospedale S. Giuseppe, Maria Letizia Bartolozzi, 2023
Centro Malattie Demielinizzanti UO Neurologia - Dipartimento di Medicina Clinica e Sperimentale - Università di Pisa - D.A.I. Neuroscienze AOUP, Livia Pasquali, 2015

TRENTINO-ALTO ADIGE/SÜDTIROL
Divisione Neurologia - Az. Spec. Ovest - Ospedale Franz Tappeiner, Francesca Caleri, 2022

UMBRIA
Centro Malattie Demielinizzanti UOC Neurologia, USL Umbria 1 Ospedale Città di Castello e Ospedale di Branca, Laura Greco, 2024
Centro Malattie Demielinizzanti - Ospedale S. Maria della Misericordia, Paola Sarchielli, 2015
Neurologia Centro SM - Ospedale San Giovanni Battista, Francesco Corea, 2015
SC di Neurofisiopatologia - AO di Perugia, Maria Grazia Celani, 2014

VENETO
Centro SM UOC Neurologia - Ospedale di Santorso, Azienda AULSS 7 - Pedemontana - Regione Veneto, Simona Carella, 2022
Fondazione Ospedale San Camillo IRCCS, Cristina Scarpazza, 2023
Centro Regionale Specializzato per la Sclerosi Multipla - UOC Neurologia B - Ospedale di Borgo Roma - AOUI Verona, Alberto Gajofatto, 2019
Reparto di Neurologia - ULSS 1 Dolomiti - Ospedale Santa Maria del Prato, Piero Nicolao, 2015
Ambulatorio Sclerosi Multipla e Malattie Demielinizzanti del SNC - UOC di Neurologia - Ospedale San Bassiano AULSS7, Alessandro Burlina, 2017
Ambulatorio Malattie Demielinizzanti - UOC di Neurologia - Ospedale di Conegliano, Marianna Fortunato, 2015
Unità di Neurologia - Centro Sclerosi Multipla - Ospedale Mater Salutis, Francesco Crescenzo, 2022
Ambulatorio Sclerosi Multipla - Divisione di Neurologia - Ospedale dell'Angelo, Rocco Quatrale, 2016
Centro Sclerosi Multipla - UOC Neurologia - Ospedale San Bortolo, Luigi Zuliani, 2022
Centro Specializzato Regionale per la Sclerosi Multipla (CeSMuV), Regione Veneto, Dipartimento di Neuroscienze DNS, Azienda Ospedaliera, Università degli Studi di Padova, Paolo Gallo, 2015
Centro Sclerosi Multipla - UOC Neurologia - Ospedale Ca' Foncello - ULSS2 Marca Trevigiana, Marika Vianello, 2015
UOC Neurologia - ULSS2 Marca Trevigiana - Ospedale S. Giacomo Apostolo, Bruno Marini, 2015